PCN30

Costs related to adverse events in chronic myeloid leukemia patients treated with tyrosine kinase inhibitors in Canada

Nick Newton PhD[1], Mireille M Goetghebeur PhD[1], Khalid El Ouagari PhD[2]
[1]*BioMedCom Consultants Inc, Montreal, QC, Canada,*
[2]*Novartis Pharmaceuticals Canada Inc., Dorval, QC, Canada.*

ABSTRACT

Background: Treatment options for the relatively small group of patients resistant or intolerant to imatinib, a recommended first-line therapy for chronic myeloid leukemia (CML), include nilotinib or dasatinib. Current data indicates that nilotinib and dasatinib have different side effect profiles.

Objectives: To investigate costs of adverse events (AEs) in patients receiving recommended doses of nilotinib or dasatinib for treatment of chronic and accelerated CML.

Methods: Incidence rates of grade 3/4 AEs in CML patients treated with nilotinib or dasatinib were obtained from clinical trial data. Direct medical costs for non-hematological AEs and for grade 4 anemia and thrombocytopenia, and febrile neutropenia were obtained from Ontario Case Costing Initiative (OCCI) inpatient databases and were specific to oncology patients. Costs for grade 3 anemia, thrombocytopenia, and non-febrile neutropenia were assumed to be outpatient costs and were based on literature and expert validation of treatment pathway and resource utilization in the Canadian context. Multivariate sensitivity analyses were conducted on costs of AEs and for an alternative dasatinib dosing (100mg).

Results: Cost of treatment-related AEs for CML patients was highest for dasatinib. Total costs for AEs associated with the accelerated phase were higher than those associated with the chronic phase: $19,902 versus $7,653 for dasatinib and $8,645 versus $3,790 for nilotinib, respectively. Ranking observed among treatments for base case costs of AEs was maintained for both high and low cost estimates, and for 100mg dasatinib dosing, indicating that the model was robust to variation in these parameters.

Conclusions: For patients resistant or intolerant to imatinib, costs of dasatinib-related AEs were approximately twice the costs of nilotinib-related AEs in both chronic and accelerated phases, highlighting the importance of considering the cost of AEs in economic evaluation of tyrosine kinase inhibitors. Further research is needed to evaluate the impact of AEs on healthcare expenditures.

INTRODUCTION

• Canadian males have a 1 in 65 probability of being diagnosed with leukemia (females 1 in 89), and a 1 in 98 probability of dying from it (females 1 in 135). Leukemia accounts for 26% of new cases (30% of deaths) due to cancer in children, and remains the most common of childhood cancers.[1]

• Economic burden of leukemia is high with mean hospital costs per patient (US data) estimated at Can $51,000.[2]

• Imatinib mesylate (Gleevec) is recommended therapy for CML treatment. Some patients treated with imatinib develop resistance or intolerance. Two treatment options are available for resistant or intolerant patients: nilotinib and dasatinib which have different side effect profiles.[3-8]

• The objective of this study was to estimate costs of adverse events (AEs) among imatinib resistant or intolerant patients receiving nilotinib or dasatinib for chronic and accelerated phases of CML.

METHODS

1 *Adverse event incidence*

• AEs were assumed to occur during the first year of treatment and were obtained from an analysis of clinical trial data.[3] Grade 3/4 AEs were considered since they are the most clinically significant and only AEs with expected significant resources utilization were included.

• Limited data was available on breakdown of grade 3 and grade 4 hematological AEs and conservative rate estimates were made based on available information (Table 1).

Table 1 Incidence rate of adverse events associated with treatment of CML

	Incidence rate (%)				
	Dasatinib 140mg[3]		Dasatinib 100mg[3]	Nilotinib[9]	
Adverse event	Chronic phase	Accelerated phase	Chronic phase	Chronic phase	Accelerated phase
Hematological events					
Anemia grade 3/4	18.0	70.0	10.0	7.9	22.9
Anemia – grade 3	*15.6**	*58.3**	*8.7**	*6.8**	*19.1**
Anemia – grade 4	*2.4**	*11.7**	*1.3**	*1.1**	*3.8**
Neutropenia	47.0	63.0	32.6§	28.3	37.1
Febrile Neutropenia	2.0	11.0	1.4§	1.3	2.5
Thrombocytopenia grade 3/4	48.0	83.0	22.0	28.5	37.4
Thrombocytopenia – grade 3	*27.4†*	*27.6†*	*12.5†*	*16.2†*	*12.3†*
Thrombocytopenia – grade 4	*20.6†*	*55.4†*	*9.5†*	*12.3†*	*25.1†*
Non-hematological events					
Gastrointestinal hemorrhage	2.0	12.0	0.9	0.9	1.7
Central nervous system hemorrhage	0.0	1.0	na	0.3	1.7
Pleural effusion	3.0	3.0	2.0	0.3	0.0
Pericardial effusion	0.9‡	1.0	0.9‡	0.3	0.0
Congestive heart failure / Cardiac dysfunction	3.0	1.0	0.0	0.3	1.7
Arrhythmia	2.0	1.0	na	0.6	0.8
Pneumonia	3.0	8.0	na	0.3	4.2
Infection	4.0	8.0	na	5.3	9.2

*breakdown of grade 3/4 anemia is an estimation based on ratios observed in imatinib prescribing information[10]; na: not available; †breakdown of grade 3/4 thrombocytopenia is an estimation based on rates observed in a phase 1 trial with dasatinib[6]; ‡product monograph states a rate of <1; §Breakdown between neutropenia (96%) and febrile neutropenia (4%) is based on percentage split observed with standard dasatinib dose (140mg).

2 *Costing of adverse events*

• Costs were estimated for one year in 2006 Canadian dollars (Can$). Non-2006 costs were inflated to 2006 using the Canadian Consumer Price Index (Table 2).

• Costs of non-hematological AEs (defined using ICD-10 codes[11]) were obtained from the Ontario Case Costing Initiative (OCCI) acute inpatient database and were specific to oncology patients as there were not enough cases in CML patients.

• Costs of hematological AEs were obtained from OCCI (inpatient data), literature and expert validation. Grade 3 and 4 hematological AEs were treated as outpatient and inpatient respectively.

Data analysis & sensitivity analyses

• The incidence and cost of AEs were combined to obtain per episode cost over one year. Calculations were performed in Microsoft Excel.

• Sensitivity analyses were performed by varying costs of AEs to obtain low and high estimates for total cost of AEs. A range of 30% was applied to the OCCI inpatient base case values (Table 2). For other costs, key assumptions were varied (e.g., number of transfusions, follow-up duration, treatment duration).

• Another sensitivity analysis was performed on AE incidence with lower dose of dasatinib since it has been reported that incidence of AEs is reduced with 100mg dasatinib dose[12,13] (however this is only applicable for chronic phase myeloid leukemia). Because limited data was available for dasatinib 100mg, only AEs for which data was available for all comparators were included.[13-15]

Table 2 Per episode costs of adverse events associated with treatment of CML

Adverse event	Cost per month or event (Can$)	Number of weeks or events	Base Case Total cost per AE (low case, high case) (Can$)	Source / Comments
Hematological events				
Anemia – grade 3	1,864.89	14 weeks	6,527.10 (4,920; 8,938)	Schedule of benefits Ontario,[16] RAMQ,[17] Manitoba cost list[18]
Anemia – grade 4	12,895	1 event	12,895 (9,027; 16,764)	OCCI
Neutropenia	135.60	3 weeks	135.60 (90; 182)	Outpatient treatment,[19] Schedule of benefits Ontario[16]
Febrile Neutropenia	37,089	1 event	37,089 (25,962; 48,216)	OCCI
Thrombocytopenia - grade 3	481.05	4 weeks	481.05 (181; 1,082)	Elting,[20] schedule of benefits Ontario[16]
Thrombocytopenia - grade 4	5,937	1 event*	5,937 (4,156; 7,718)	OCCI, Elting,[20] schedule of benefits Ontario[16]
Non-hematological events				
Gastrointestinal hemorrhage	12,309	1 event	12,309 (8,616; 16,002)	OCCI
Central nervous system hemorrhage	21,451	1 event	21,451 (15,016; 27,886)	OCCI†
Pleural effusion	25,480	1 event	25,480 (17,836; 33,124)	OCCI
Pericardial effusion	17,477	1 event	17,477 (12,234; 22,720)	OCCI
Congestive heart failure / Cardiac dysfunction	22,097	1 event	22,097 (15,468; 28,726)	OCCI
Arrhythmia	33,588	1 event	33,588 (23,512; 43,664)	OCCI
Pneumonia	24,448	1 event	24,448 (17,114, 31,782)	OCCI
Infection	23,219	1 event	23,219 (16,253; 30,185)	OCCI‡

OCCI: Ontario Case Costing Initiative; RAMQ: Régie de l'assurance maladie du Québec.
*assumed half of patients would be hospitalized (OCCI cost) and other half would incur cost of high case for grade 3 thrombocytopenia
†OCCI cost for all patient types; cost in oncology patients not available due to too few cases
‡cost is for urinary tract infection, cost for code B99 not available due to less than 5 cases

RESULTS

1 *Costs related to chronic phase myeloid leukemia*

• Overall cost of treatment-related AEs was highest for dasatinib ($7,653) compared to nilotinib ($3,790).

• Thrombocytopenia, anemia, infection and pleural effusion were biggest cost drivers for dasatinib treatment.

• Infection, thrombocytopenia, febrile neutropenia and anemia were biggest cost drivers for nilotinib treatment.

Figure 1 Per patient costs of adverse events related to treatment of chronic phase myeloid leukemia

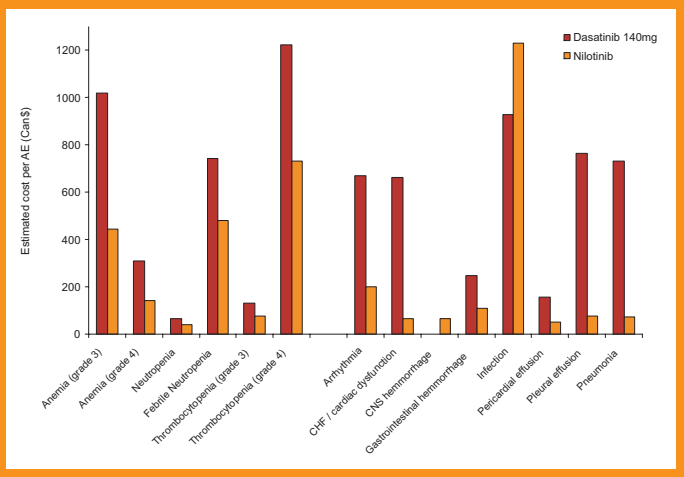
Dasatinib 140mg
Nilotinib
Estimated cost per AE (Can$)
0
200
400
600
800
1000
1200
Anemia (grade 3)
Anemia (grade 4)
Neutropenia
Febrile Neutropenia
Thrombocytopenia (grade 3)
Thrombocytopenia (grade 4)
Arrhythmia
CHF / cardiac dysfunction
CNS hemmorrhage
Gastrointestinal hemmorrhage
Infection
Pericardial effusion
Pleural effusion
Pneumonia

2 *Costs related to accelerated phase myeloid leukemia*

• Total costs for AEs with accelerated phase were significantly higher than costs associated with chronic phase; $19,902 compared with $7,653 for dasatinib and $8,645 compared with $3,790 for nilotinib.

• Average per patient cost of AEs was higher for dasatinib ($19,902), than for nilotinib ($8,645).

• Febrile Neutropenia, anemia and thrombocytopenia contributed most to dasatinib AE costs, while infection, thrombocytopenia and anemia were greatest contributors to nilotinib AE costs.

Figure 2 Per patient costs of adverse events related to treatment of accelerated phase myeloid leukemia

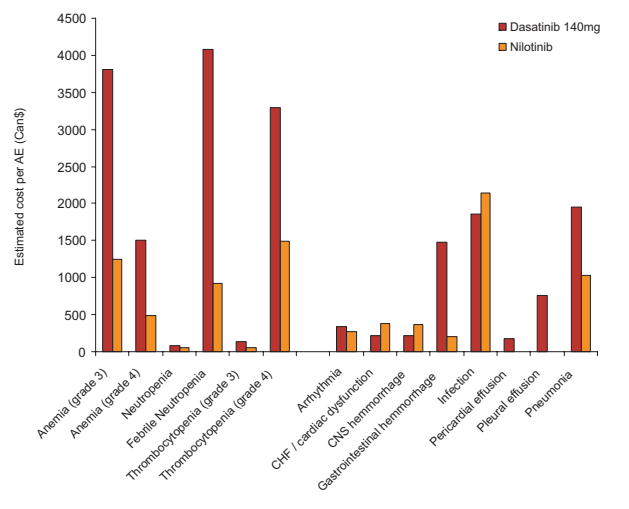
Dasatinib 140mg
Nilotinib
Estimated cost per AE (Can$)
0
500
1000
1500
2000
2500
3000
3500
4000
4500
Anemia (grade 3)
Anemia (grade 4)
Neutropenia
Febrile Neutropenia
Thrombocytopenia (grade 3)
Thrombocytopenia (grade 4)
Arrhythmia
CHF / cardiac dysfunction
CNS hemmorrhage
Gastrointestinal hemmorrhage
Infection
Pericardial effusion
Pleural effusion
Pneumonia

3 *Sensitivity analysis*

• For chronic phase CML, range in overall costs of treating adverse events was larger for dasatinib than for nilotinib, and for accelerated phase CML the range in overall costs of treating adverse events was also larger with dasatinib than for nilotinib (figure 3).
• Where costs of adverse events were varied using the low and high case costs (Table 2) then the ranking observed among treatments for base case costs of AEs was maintained for both the high and low cost estimates (figure 3).

Figure 3 Costs resulting from variation of annual per patient cost of adverse events related to treatment of chronic and accelerated phase myeloid leukemia

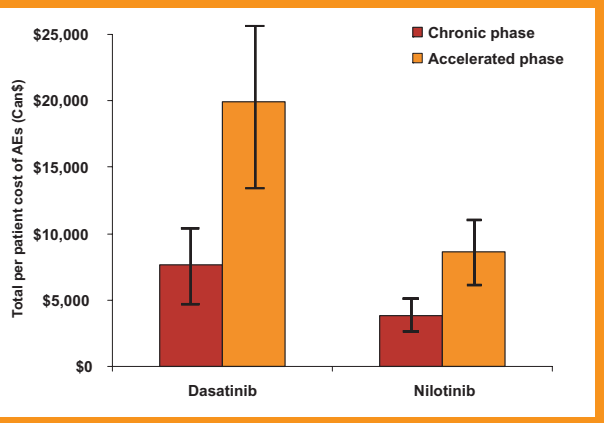
Chronic phase
Accelerated phase
Total per patient cost of AEs (Can$)
$0
$5,000
$10,000
$15,000
$20,000
$25,000
Dasatinib
Nilotinib

• Where incidence of adverse events was varied, using the 100mg dasatinib dose, the largest adverse event cost burden was associated with 140mg dasatinib followed by 100mg dasatinib and nilotinib (Table 3). Note this is only applicable to the chronic phase since dose reduction is not permissible with accelerated phase.

Table 3 Costs resulting from variation of incidence of AE with reduced dasatinib dose related to treatment of chronic phase myeloid leukemia

	Annual cost per patient (Can$)		
Adverse event	**Dasatinib 140mg**	**Dasatinib 100mg**	**Nilotinib**
Hematological adverse events			
Anemia (grade 3)	1,018	568	444
Anemia (grade 4)	309	168	142
Neutropenia	64	44	38
Febrile Neutropenia	742	519	482
Thrombocytopenia (grade 3)	132	60	78
Thrombocytopenia (grade 4)	1,223	564	730
Non-hematological adverse events			
CHF / cardiac dysfunction	663	0	66
Gastrointestinal hemmorrhage	246	111	111
Pericardial effusion	157	157	52
Pleural effusion	764	510	76
Total	**5,318**	**2,701**	**2,153**

LIMITATIONS

• Breakdown of grade 3/4 hematological AEs was estimated based on available data. Although actual rate values may differ, a conservative approach was taken and the same breakdown was applied for both tyrosine kinase inhibitors.

• Treatment duration for anemia was based on studies with oncology patients, which usually refers to chemotherapy-induced anemia. Chemotherapy-related hematological AEs are stringent and generally stop after chemotherapy is completed. Anemia associated with a chronic treatment is likely to be more persistent and treatment duration used in this model may have underestimated actual cost of anemia.

• In case of persistent treatment-related AEs, the patient may also be switched to another treatment, which is not considered in this model.

CONCLUSIONS

• For patients resistant or intolerant to imatinib, costs of dasatinib-related AEs were approximately twice the costs of nilotinib-related AEs in both chronic and accelerated phases.

• Ranking observed among treatments for base case total costs of AEs was maintained for both high and low cost estimates and for reduced (100mg) dasatinib dosage, indicating that the model was robust to variation in these parameters.

• This study highlights the importance of considering adverse events in the economic assessment of treatment of CML patients with tyrosine kinase inhibitors.

ACKNOWLEDGEMENTS

We would like to thank Dr Denis Cournoyer from the Montreal General Hospital Research Institute, Montreal, Canada for expert validation.

REFERENCES

1. Canadian Cancer Society, National Cancer Institute of Canada, Statistics Canada, Provincial/Territorial Cancer Registries, Public Health Agency of Canada. Canadian cancer statistics 2006. 2006. www.cancer.ca. (Accessed 29 Sep 2006). 2. Kasteng F, Sobocki P, Svedman C, Lundkvist J. Economic evaluations of leukemia: A review of the literature. Int J Technol Assess Health Care. 2007;23(1):43-53. 3. BMS. Dasatinib: BMS 354825. Drugs R D. 2006;7(2):129-32. 4. Novartis. Tasigna product monograph. 2006. 5. Kantarjian H, Giles F, Wunderle L, Bhalla K, O'brien S, Wassmann B, et al. Nilotinib in imatinib-resistant CML and Philadelphia chromosome-positive ALL. N Engl J Med. 2006;354(24):2542-51. 6. Talpaz M, Shah NP, Kantarjian H, Donato N, Nicoll J, Paquette R, et al. Dasatinib in imatinib-resistant Philadelphia chromosome-positive leukemias. N Engl J Med. 2006;354(24):2531-41. 7. Cortes J, Kim DW, Guilhot F, Rosti G, Silver RT, Gollerkeri A, et al. Dasatinib (SPRYCEL) in Patients (pts) with Chronic Myelogenous Leukemia in Accelerated Phase (AP-CML) That Is Imatinib-Resistant (im-r) or -Intolerant (im-i): Updated Results of the CA180-005 START-A Phase II Study. Blood 2006.108:(11)A-2160 8. Kantarjian H, Ottmann O, Gattermann N, Larson R, Rafferty T, Alland L, et al. A Phase II study of nilotinib (AMN107) a novel inhibitor of bcr-abl, administered to imatinib-resistant or intolerant patients with chronic myelogenous leukemia (CML) in accelerated phase (AP); Abstract presented at ASCO; 2006. Proceedings of the 11th Congress of the European Hematology Association 2006.June 15-18:(Amsterdam)A-0156 9. Novartis Pharmaceuticals Canada I. Summary of clinical safety- 120 day safety update. 2007. 10. Novartis Pharmaceuticals Canada I. Gleevec prescribing information 23May2007. 2007. 11. Canadian Institute for health information. International statistical classification of diseases and related health problems, 10th revision (ICD-10). 2006. 12. Bristol-Myers Squibb. Sprycel product monograph. 2006. 13. Bristol-Myers Squibb. Sprycel: highlights of prescribing information. 2007. 14. Shah, N. P. Dasatinib 50 mg or 70 mg BID compared to 100 mg or 140 mg QD in patients with CML in chronic phase (CP) who are resistant or intolerant to imatinib: One-year results of CA180034. Abstract presented at 43th ASCO Annual Meeting; 2007 Jun 1-5. Chicago; 2007. 15. Hochhaus, A. Dasatinib dose and schedule optimization in chronic-phase cml resistant or intolerant to imatinib: results from a randomized phase-III trial (CA180034). Abstract presented at 12th Congress of the European Hematology Association; 2007 Jun 7-10. Vienna; 2007. 16. Ontario Ministry of Health and Long-Term Care. Ontario drug benefit formulary/comparative index: electronic version. 2006. http://www.health.gov.on.ca/english/providers/program/drugs/odbf_eformulary.html. (Accessed 24 Apr 2007). 17. Regie de l'assurance maladie du Québec. Liste de médicaments assurés édition 15 modification 20. 2007. http://www.ramq.gouv.qc.ca/fr/professionnels/listmed/lm_tdmf.shtml. (Accessed 23 Feb 2007). 18. Manitoba Centre for Health Policy. Appendix 1 & 2 from cost list for Manitoba health services. 2003. http://www.umanitoba.ca/centres/mchp/concept/dict/costing/hospital_costing.html. 19. Berkow R and Fletcher A, (ed.). The Merck manual of diagnosis and therapy. 15th ed. Rahway,NJ: Merck Sharp & Dohme Research Laboratories, Division of Merck & Co. Inc.;1987. 20. Elting LS, Cantor SB, Martin CG, Hamblin L, Kurtin D, Rivera E, et al. Cost of chemotherapy-induced thrombocytopenia among patients with lymphoma or solid tumors. Cancer. 2003;97(6):1541-50.