Economic evaluation of intensive atorvastatin treatment versus standard simvastatin treatment in Canada based on the IDEAL trial

M. Wagner[1], P. Lindgren[2], E. Merikle[3], M. Goetghebeur[1], B. Jönsson[2], & the IDEAL Steering Committee

[1]BioMedCom Consultants inc, Montreal, QC, Canada; [2]European Health Economics, Stockholm, Sweden; [3]Pfizer Canada Inc., Montreal, QC, Canada

Background

- The IDEAL trial—a prospective, randomized, open-label, blinded endpoint trial—compared treatment with high-dose atorvastatin (80 mg/day) to usual-dose simvastatin (20–40 mg/day) in 8888 patients from Northern Europe who had a history of acute myocardial infarction (MI) over a median follow-up of 4.8 years[1]
- Mean LDL-C levels decreased from 3.15 mmol/L at baseline to 2.69 mmol/L and 2.10 mmol/L in simvastatin and atorvastatin patients, respectively.[1] Atorvastatin patients experienced fewer major coronary events (hazard ratio [HR], 0.89; 95% CI, 0.78–1.01, *P*=.07) and significantly fewer coronary heart disease (HR 0.84, 95% CI 0.76–0.91, *P*<.001) and cardiovascular events (HR 0.84, 95% CI 0.78–0.91, *P*<.001)[1]
- A trial-based economic evaluation indicated that atorvastatin 80 mg would be cost-effective compared to simvastatin 20–40 mg in Sweden and the USA.[2] Given the availability of generic simvastatin in Canada at an acquisition cost significantly lower than that of atorvastatin, the question is whether treatment with high-dose atorvastatin would represent a cost-effective use of healthcare resources in Canada

Objective

To assess from a Canadian societal prospective the cost-effectiveness of intensive lipid lowering with high-dose atorvastatin (80 mg/day) versus moderate lipid lowering with standard dose generic simvastatin (20–40 mg/day) in patients with a history of MI, based on IDEAL trial data

Methods

- **Within-trial analysis**: endpoint-related events, resources used (i.e., endpoint-related hospitalization and study drugs) and productivity losses occurring during the IDEAL trial were aggregated by treatment arm on an intention-to-treat basis. *Cost-effectiveness* was calculated as the incremental cost per event avoided
 - *Endpoint-related hospitalization:* hospital stay with a diagnosis of stroke, transient ischemic attack, non-fatal MI, heart failure, cardiac arrest, or angina or for heart transplantation, coronary bypass surgery, percutaneous coronary intervention or another cardiothoracic or vascular procedure. Each hospitalization was assigned a cost (including overhead) based on Ontario Cost Case Initiative 2003-2004 acute inpatient cost data[3] (Table 1)
 - *Study drugs*: atorvastatin arm: 80-mg dose used on 90.7% of study days and 40 mg on 9.3%; simvastatin arm: 20-mg dose used on 79.3% of study days and 40 mg on 20.7%. Drug costs were obtained from the current Quebec formulary[4]
 - *Productivity loss*: Estimated at Can$901.21 per week based on average weekly earnings[5] and a 20% employer contribution
- Additionally, a **lifetime Markov model** with 1-year cycle length was developed (Figure 1).[6] *Transition probabilities* were based on patient-level data from the IDEAL trial (Table 2). After five years, patients in both arms were assumed to have the same risks. *Cost-effectiveness* was calculated as incremental cost per quality-adjusted life-year (QALY)
 - *Baseline patient characteristics:* the initial age of the cohort was assumed to be 62 years, the baseline LDL-C 2.75 mmol/L, the proportion of women 30%, and the proportion of patients with diabetes 25% based on Canadian studies[7-10]
 - Costs and benefits were *discounted* at 5%
 - *Utilities*: Based on published studies (Table 3)[11-12]

Table 1: Mean number of hospitalizations during the IDEAL study and corresponding Canadian costs

Diagnosis or procedure (NordDRG code)	Mean number of hospitalizations per patient		Canadian cost (2006 Can$)*
	Atorvastatin 80 mg/d arm	Simvastatin 20–40 mg/d arm	
Stroke (014)	0.035	0.045	16,724
TIA (015)	0.013	0.013	6,027
Non-fatal MI (121, 122)	0.054†	0.052†	11,444
Heart failure (127)	0.032	0.038	9,835
Angina pectoris (140)	0.038	0.044	4,499
Cardiac arrest (129)	0.002	0.002	17,934
PCI (112)	0.097	0.127	8,856
CABG (106, 107)	0.052	0.069	21,603
Cardiac transplant (103)	0	0.001	141,893
Other cardiothoracic procedure (108)	0	0.001	30,018‡
Other vascular procedure (478, 479)	0.028	0.040	8,856§

CABG: coronary artery bypass graft; DRG: Diagnosis Related Group; MI: myocardial infarction; PCI: percutaneous coronary intervention; TIA: transient ischemic attack *Source: Ontario Case Costing Initiative;[3] † MI without a procedure performed during the same hospitalization; including all MI hospitalizations, the total number was 0.08 per patient in the atorvastatin and 0.09 in the simvastatin arm; ‡ assumed to be 1.39 times the cost of CABG; § assumed to equal the cost of PCI

Figure 1: Lifetime Markov model

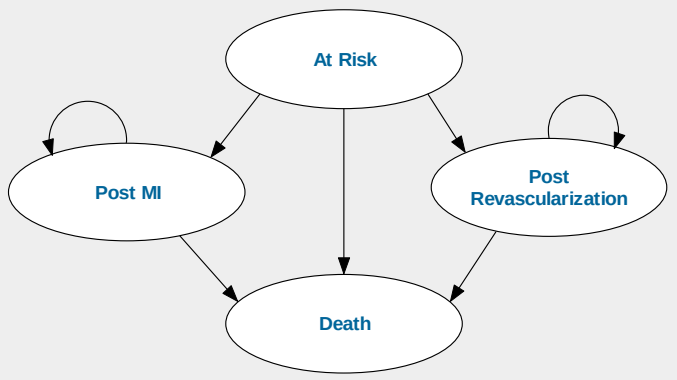
At Risk
Post MI
Post Revascularization
Death

Table 2: Coefficients from Weibull regression models used to calculate Markov transition probabilities

Parameter	Coefficient (+/-standard error)			
	Risk of MI	Risk of revascularization	Mortality without event	Mortality after MI
Baseline characteristics				
Age	0.015 (0.006)	-0.019 (0.004)	0.068 (0.005)	0.055 (0.013)
Female	NS	NS	-0.314 (0.111)	NS
Diabetes	0.481 (0.156)	NS	0.338 (0.115)	0.420 (0.220)
LDL-C (mg/dL)	0.005 (0.002)	0.003 (0.001)	0.004 (0.001)	NS
Weibull regression parameters				
Constant	-9.856 (0.540)	-5.120 (0.334)	-15.833 (0.485)	-10.302 (0.989)
Shape parameter	0.754 (0.044)	0.535 (0.022)	1.105 (0.046)	0.685 (0.055)

LDL-C: low-density lipoprotein cholesterol; MI: myocardial infarction; NS: not significant for this outcome
Source: Patient-level IDEAL trial data

Table 3: Health utilities applied to the Markov model

	Utility value (+/-standard deviation)	
Age (years)	*Female**	*Male**
50	0.82 (0.012)	0.84 (0.013)
60	0.78 (0.015)	0.83 (0.012)
70	0.78 (0.017)	0.81 (0.017)
80	0.74 (0.026)	0.74 (0.026)
Event	*Decrement applied during first year after event†*	
MI	0.05 (0.01)	
Revascularization	0.03 (0.01)	

Source: *Burstrom et al., 2001;[11] †Lindgren et al., 2006[12]

Results

1 *Within-trial cost effectiveness*

- Over 4.8 years of treatment, study drug costs per patient were on average Can$968 higher in the atorvastatin than in the simvastatin arm. However, this was more than offset by lower costs for endpoint-related hospital stays and lower productivity losses, resulting in a net cost saving of Can$663 per patient in the atorvastatin arm
- Patients in the atorvastatin arm experienced on average 0.099 fewer events over the trial period than patients in the simvastatin arm
- Since patients in the atorvastatin arm incurred fewer costs and had fewer events, atorvastatin treatment was dominant

	Atorvastatin (80 mg/day)	Simvastatin (20–40 mg/day)	Difference
Costs (Can$)			
Study drug	3,171	2,203	968
Endpoint-related hospitalization	4,039	5,109	-1,070
Productivity loss	20,154	20,714	-561
Total	27,364	28,026	-663
Average number of events per patient	0.462	0.561	0.099
Total incremental cost per event avoided (Can$) [95% CI]		Dominant [dominant to 1,414]	
Direct costs only (Can$) [95%CI]		Dominant [dominant to 5,408]	

All costs are expressed in 2006 Canadian dollars; CI: confidence interval

2 *Probabilistic sensitivity analysis (bootstrapping)*

- The figure below shows the total incremental cost of atorvastatin treatment versus number of events avoided over 4.8 years of treatment based on 1000 bootstrapping replications
- All replications indicated fewer events with atorvastatin 80 mg (i.e., all points are located to the right of the y-axis)
- 72.6% of replications indicate cost savings with atorvastatin 80 mg (i.e., located below the x-axis)
- In over 88% of replications the total incremental cost per event avoided was below Can$7000

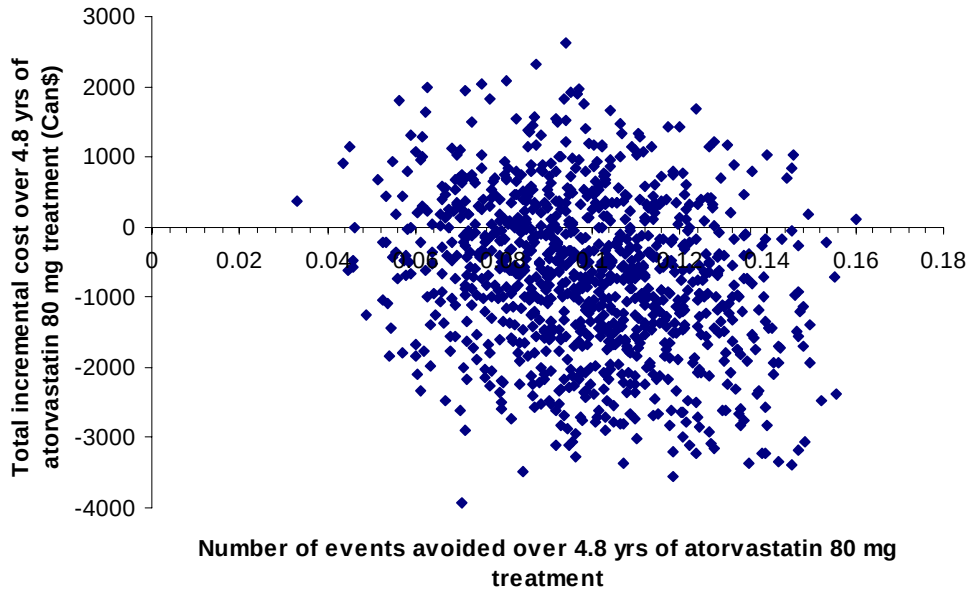
3000
2000
1000
0
-1000
-2000
-3000
-4000
0
0.02
0.04
0.06
0.08
0.1
0.12
0.14
0.16
0.18
Total incremental cost over 4.8 yrs of atorvastatin 80 mg treatment (Can$)
Number of events avoided over 4.8 yrs of atorvastatin 80 mg treatment

3 *Lifetime Markov model results*

- Patients in the atorvastatin arm were projected to live on average 0.036 years longer and have 0.023 more QALYs than patients in the simvastatin arm. This improvement came at a total incremental cost of Can$206 per patient. The resulting incremental cost-effectiveness ratio (ICER) was Can$5,768 per life-year gained or Can$8,808 per QALY gained
- Exclusion of indirect costs (productivity losses) resulted in an ICER of Can$13,664 per life-year or Can$20,866 per QALY gained

	Atorvastatin (80 mg/day)	Simvastatin (20–40 mg/day)	Difference
Projected health outcomes per patient			
Life-years	11.553	11.517	0.036
QALYs	8.718	8.742	0.023
Projected costs per patient, Can$			
Study drug	5,316	4,362	954
Endpoint-related hospitalization	4,137	4,603	-466
Productivity loss	1,071	1,353	-282
Total	10,524	10,318	206
Incremental cost-effectiveness ratios, Can$			
Cost per LYG		5,768	
Cost per QALY gained		8,808	

CI: confidence interval; LYG: life-year gained; QALY: quality-adjusted life year

Conclusions

- The within-trial cost-effectiveness analysis indicates that treatment of patients who have a history of MI with atorvastatin 80 mg versus simvastatin 20-40 mg is cost-saving over a median of 4.8 years from a Canadian societal perspective
- Atorvastatin was cost saving in 72.6% of 1000 bootstrapping replications; in over 88% of replications the total incremental cost per event avoided was below Can$7000
- The Markov model suggests that treatment with atorvastatin 80 mg would be cost-effective over a lifetime horizon at Can$5,768 per life-year gained or Can$8,808 per QALY gained

References

1 Pedersen TR, Faergeman O, Kastelein JJ, Olsson AG, Tikkanen MJ, Holme I, et al. High-dose atorvastatin vs usual-dose simvastatin for secondary prevention after myocardial infarction: the IDEAL study: a randomized controlled trial. JAMA. 2005;294(19):2437-45. ***2*** Lindgren P, Jonsson B. Trial based economic evaluation of intensive atorvastatin compared to standard simvastatin in the IDEAL trial. 65th Annual Conference of the AHA; 2006 May 11-14; Tampa, FL. ***3*** Ontario Ministry of Health and Long-Term Care. Ontario Case Costing Initiative. On Ontario Case Costing Initiative website [updated 2004; cited 2006 Jul 5]. Available from: http://www.occp.com/. ***4*** Régie de l'assurance maladie du Québec. Liste de médicaments assurés. 2007 Feb 7. ***5*** Statistics Canada. Earnings, average weekly, by province and territory. On Statistics Canada website [updated 2006 Jun 30; cited 2006 Aug 30]. Available from: http://www40.statcan.ca/l01/cst01/-labr79.htm. ***6*** Lindgren P, Graff J, Olsson AG, Pedersen TJ, Jonsson B. Cost-effectiveness of high-dose atorvastatin compared with regular dose simvastatin. Eur Heart J. 2007; published online on March 19th. ***7*** Bata IR, Gregor RD, Wolf HK, Brownell B. Trends in five-year survival of patients discharged after acute myocardial infarction. Can J Cardiol. 2006;22(5):399-404. ***8*** Yan RT, Yan AT, Tan M, Chow CM, Fitchett DH, Ervin FL, et al. Age-related differences in the management and outcome of patients with acute coronary syndromes. Am Heart J. 2006;151(2):352-9. ***9*** Alter DA, Chong A, Austin PC, Mustard C, Iron K, Williams JI, et al. Socioeconomic status and mortality after acute myocardial infarction. Ann Intern Med. 2006;144(2):82-93. ***10*** Petrella RJ, Merikle E, Jones J. Prevalence and treatment of dyslipidemia in Canadian primary care: a retrospective cohort analysis. Clin Ther. 2007;29(4):742-50. ***11*** Burstrom K, Johannesson M, Diderichsen F. Swedish population health-related quality of life results using the EQ-5D. Qual Life Res. 2001;10(7):621-35. ***12*** Lindgren P, Stenestrand U, Hambraeus K, Wallentin L, Jonsson B. PCI reduces utility loss after MI in Sweden. Eur Heart J. 2006;27(Suppl 1):773.